682

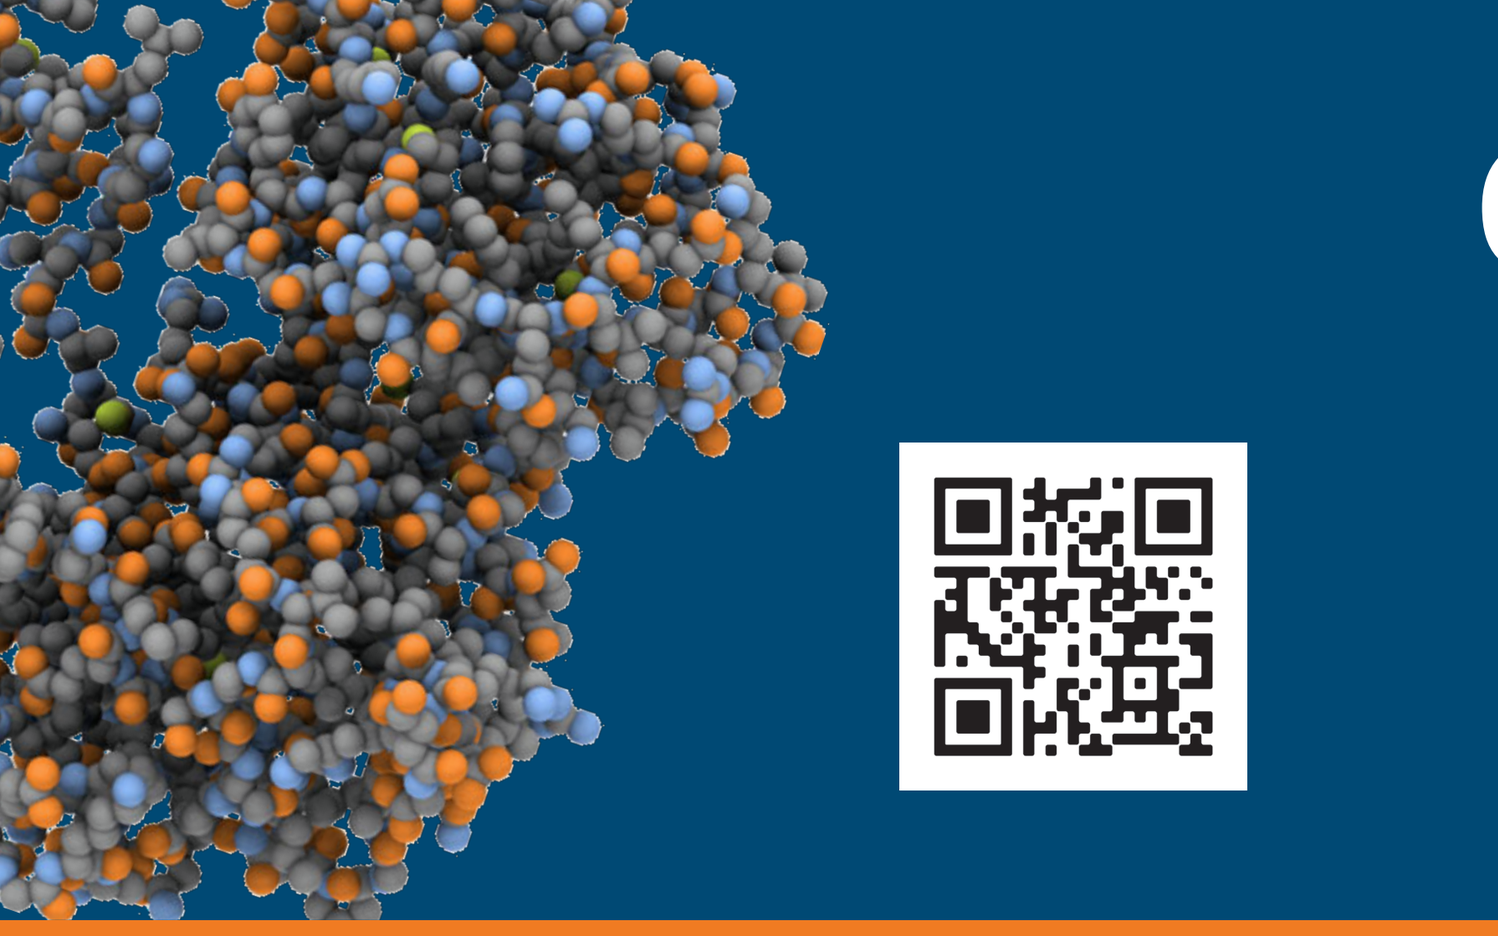

Ongoing Phase 1/2 Trial of the HPK1 Inhibitor NDI-101150 as Monotherapy and in Combination with Pembrolizumab: Clinical Safety Update and Renal Cell Carcinoma (RCC) Efficacy Analysis

David Sommerhalder[1a]; Kurt Demel[2]; Marcus S. Noel[3]; Hamid Emamekhoo[4]; Julio Peguero[5]; Rama Balaraman[6]; Ryan H. Moy[7]; Anita Scheuber[8]; Joanna Haas[8]; Daria Chabas[8]; Pavan Kumar[8]; Scott Daigle[8]; Sue Dasen[8]; Xinyan Zhang[8]; Martin Gutierrez[9]; Sunil Sharma[10]; Arif Hussain[11]

[1]NEXT Oncology, San Antonio, TX, USA; [2]HealthPartners, St. Louis Park, MN, USA; [3]Georgetown University Medical Center – Lombardi Comprehensive Cancer Center, Washington, DC, USA; [4]University of Wisconsin Carbone Cancer Center, Madison, WI, USA; [5]Oncology Consultants, Houston, TX, USA; [6]Florida Cancer Affiliates-US Oncology, Ocala, FL, USA; [7]Columbia University Irving Medical Center, New York, NY, USA; [8]Nimbus Therapeutics, Boston, MA, USA; [9]Hackensack University Medical Center – John Theurer Cancer Center, Hackensack, NJ, USA; [10]Honor Health Research Institute, Scottsdale, AZ, USA; [11]University of Maryland Greenebaum Comprehensive Cancer Center, Baltimore, MD, USA

[a]Presenting author: David Sommerhalder (dsommerhalder@nextoncology.com)

BACKGROUND AND METHODS

- NDI-101150 is a potent, selective, oral inhibitor of hematopoietic progenitor kinase 1 (HPK1; **Fig. 1**). Previous analyses have shown an acceptable safety profile and preliminary evidence of efficacy in patients with advanced solid tumors[1–4]
- NDI-101150 is currently being investigated in a multicenter, open-label, phase 1/2 trial (NCT05128487) as monotherapy (50–200 mg once daily [QD]) or in combination with pembrolizumab (NDI-101150: 50 or 100 QD; pembrolizumab: 200 mg QD) in patients with advanced solid tumors (**Fig. 2**)
- We report here updated safety data in all dose cohorts and tumor types as well as efficacy data in patients with renal cell carcinoma (RCC) receiving NDI-101150 monotherapy

Figure 1. HPK1 is a compelling immuno-oncology target

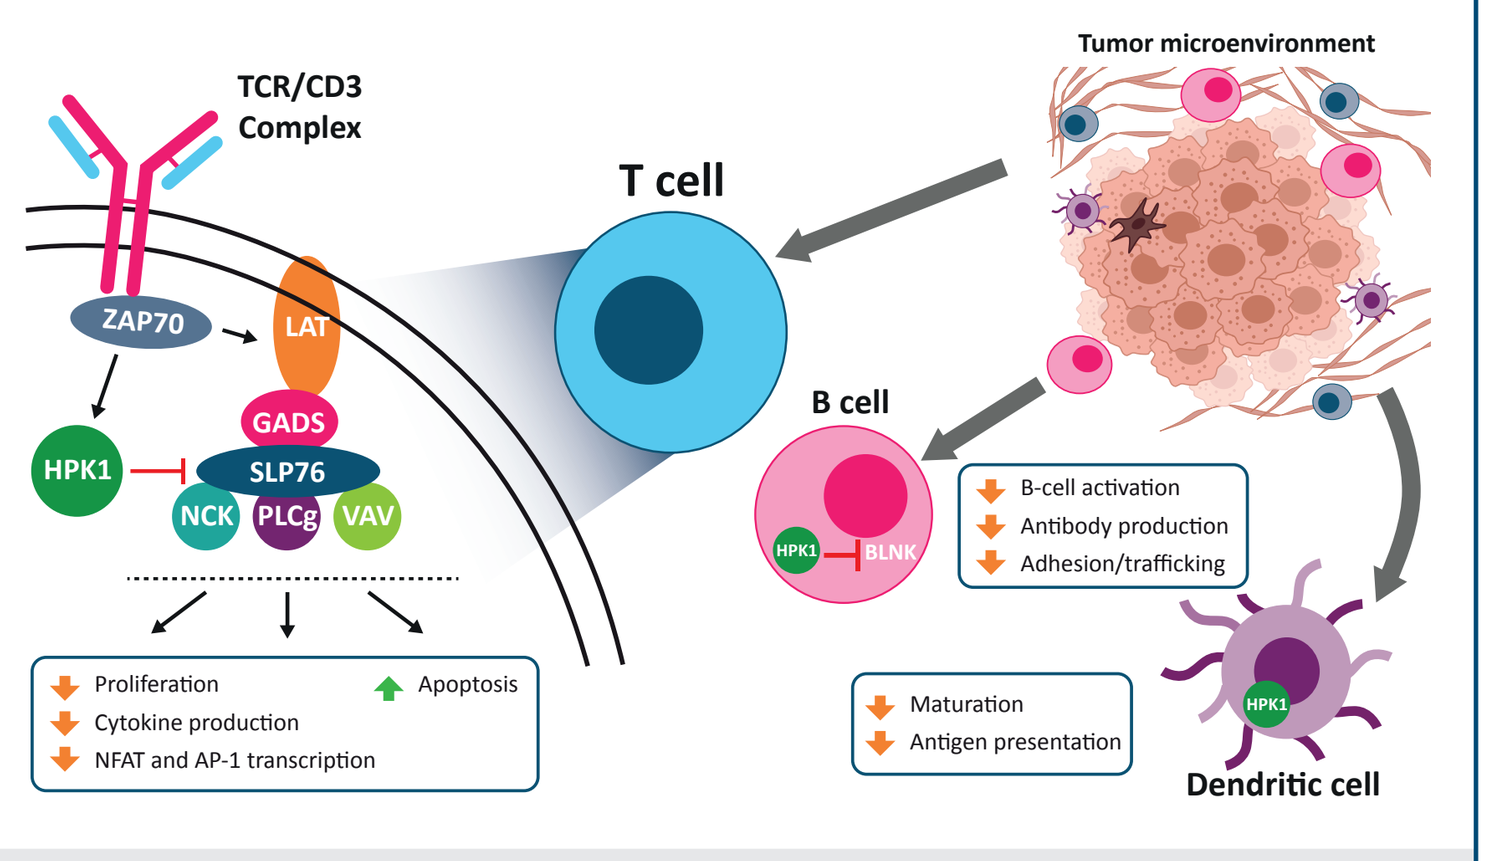

TCR/CD3 Complex
T cell
Tumor microenvironment
ZAP70
LAT
GADS
HPK1
SLP76
NCK
PLCg
VAV
B cell
HPK1
BLNK
B-cell activation
Antibody production
Adhesion/trafficking
Proliferation
Cytokine production
NFAT and AP-1 transcription
Apoptosis
Maturation
Antigen presentation
Dendritic cell

AP-1, activator protein 1; BLNK, B-cell linker protein; GADS, GRB2 related adaptor protein downstream of Shc; HPK1, hematopoietic progenitor kinase 1; LAT, linker for activation of T cells; NCK, non-catalytic region of the tyrosine kinase; NFAT, nuclear factor of activated T cells; PLCg, phospholipase C, gamma 1; SLP76, SH2 domain containing leukocyte protein of 76kDa; TCR, T cell receptor; ZAP70, zeta-chain-associated protein kinase 70

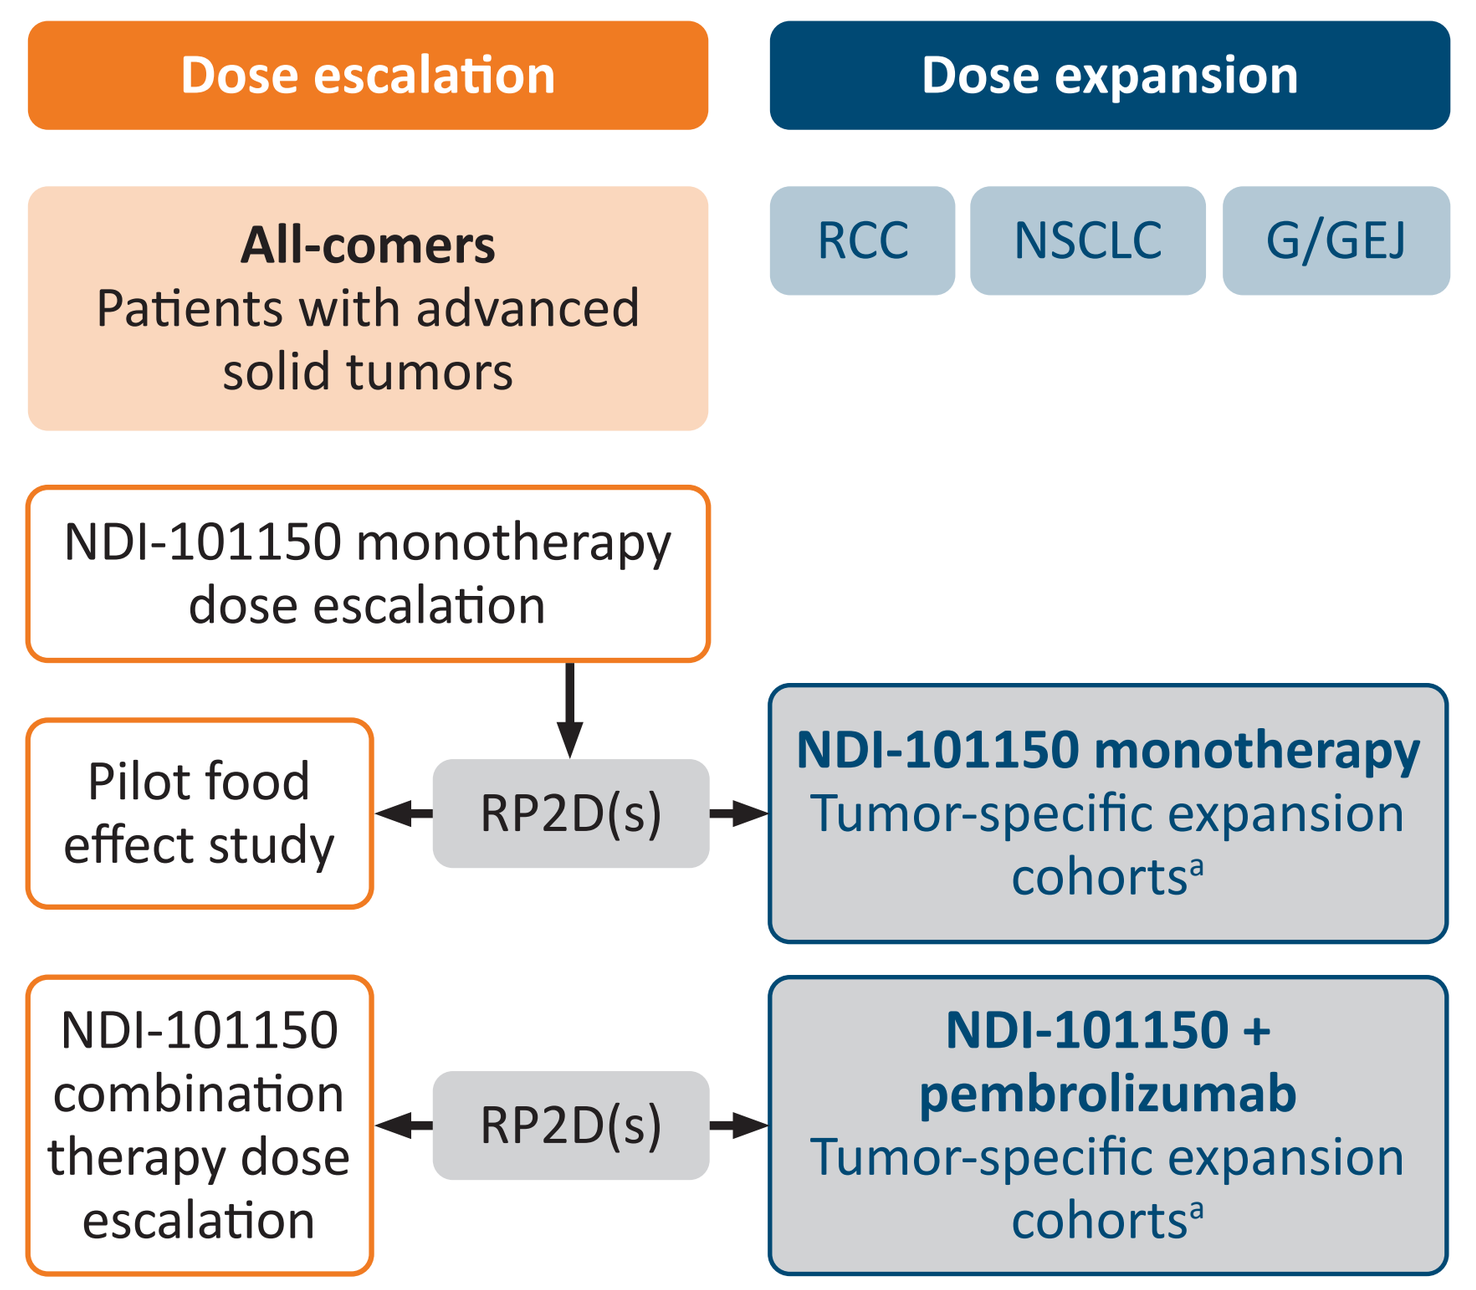

Dose escalation
Dose expansion
All-comers
Patients with advanced solid tumors
RCC
NSCLC
G/GEJ
NDI-101150 monotherapy dose escalation
Pilot food effect study
RP2D(s)
NDI-101150 monotherapy
Tumor-specific expansion cohorts[a]
NDI-101150 combination therapy dose escalation
RP2D(s)
NDI-101150 + pembrolizumab
Tumor-specific expansion cohorts[a]

[a]Response triggers opening of additional tumor-specific cohorts
G/GEJ, gastric/gastro-esophageal junction cancer; NSCLC, non-small cell lung cancer; RCC, renal cell carcinoma; RP2D, recommended phase 2 dose

RESULTS

- As of 12 August 2024 (data cut off), 53 patients had been dosed in the dose escalation cohorts (41 receiving NDI-101150 monotherapy and 12 receiving NDI-101150 + pembrolizumab) and 35 patients had been dosed in the NDI-101150 monotherapy dose expansion cohorts
- The safety analysis set comprised all 88 patients and the efficacy analysis set was comprised of 17 patients (all of whom were patients with RCC and ≥1 post-baseline assessment and who received NDI-101150 monotherapy)

Demographics

- Median age was 66 years (range: 21–87) and 57% of patients were males; the most common tumor type was RCC (n=24; 27%), followed by non-small cell lung cancer (n=14; 16%), gastric/gastro-esophageal junction (n=10; 11%), and colorectal cancer (n=7; 8%)
- In the safety analysis set, the median number of prior treatments was 3 (range: 1–10); in the RCC cohort (efficacy analysis set), the median number of prior treatments was 3 (range: 1–7)

Safety

- The most common adverse events (any grade) considered related to NDI-101150 (TRAEs) were nausea (n=36; 41% of patients), diarrhea (n=30; 34%), vomiting (n=27; 31%), and fatigue (n=23; 26%) (**Table 1**)
- Grade 3 TRAEs occurred in 12 (14%) patients and included fatigue (n=2; 2%), and one each (n=1; 1% of patients) of diarrhea, constipation, colitis, hypersensitivity, aspartate aminotransferase increased, platelet count decreased, hypokalemia, acute kidney injury, proteinuria, dyspnea, hypoxia, immune-mediated lung disease, pneumonitis, and nephritis; one patient (1%) experienced a grade 4 TRAE (aplastic anemia); all other TRAEs were grade 1 or 2
- The most frequent immune-related (IR) TRAEs of any grade, as determined by the investigator, were diarrhea (n=6; 7%), vomiting (n=4; 5%), rash (n=4; 5%), colitis (n=3; 3%), and nausea (n=3; 3%) (**Table 2**)
- Eight (9%) patients experienced at least one grade ≥3 IR-TRAE, most of whom were receiving NDI-101150 200 mg (n=3; 33%). These IR-TRAEs occurred in one patient each (n=1; 1% of patients) and were as follows: acute kidney injury, aplastic anemia, elevated aspartate aminotransferase, colitis, hypersensitivity, hypoxia, immune-mediated lung disease, platelet count decreased, pneumonitis, and nephritis

Table 1. Any-grade TRAEs occurring in ≥4 patients in the safety analysis set (N=88)[a]

TRAEs, n (% patients)	NDI-101150 monotherapy (n=76)					Combination therapy[b] (n=12)		
	50 mg (n=8)	100 mg[c] (n=36)	140 mg (n=5)	150 mg[c] (n=18)	200 mg (n=9)	50 mg (n=8)	100 mg (n=4)	Overall (n=88)
≥1 TRAE	8 (100)	31 (86)	3 (60)	9 (50)	8 (90)	8 (100)	3 (75)	70 (80)
Preferred term								
Nausea	6 (75)	16 (44)	1 (20)	6 (33)	3 (33)	2 (25)	2 (50)	36 (41)
Diarrhea	1 (13)	15 (42)	2 (40)	2 (11)	6 (67)	2 (25)	2 (50)	30 (34)
Vomiting	3 (38)	11 (31)	0	5 (28)	5 (56)	2 (25)	1 (25)	27 (31)
Fatigue	2 (25)	13 (36)	2 (40)	1 (6)	3 (33)	1 (13)	1 (25)	23 (26)
Anemia	1 (13)	3 (8)	0	2 (11)	1 (11)	0	0	7 (8)
Blood creatinine increased	0	5 (14)	0	0	0	0	0	5 (6)
Constipation	0	2 (6)	0	0	1 (11)	1 (13)	1 (25)	5 (6)
Platelet count decreased	1 (13)	2 (6)	0	1 (6)	1 (11)	0	0	5 (6)
Pruritus	1 (13)	3 (8)	0	0	1 (11)	0	0	5 (6)
Abdominal pain	1 (13)	0	0	1 (6)	1 (11)	0	1 (25)	4 (5)
Decreased appetite	0	2 (6)	1 (20)	1 (6)	0	0	0	4 (5)
Rash[d]	2 (25)	0	1 (20)	0	1 (11)	0	0	4 (5)

[a]Patients reporting more than one event are counted only once for each preferred term; [b]NDI-101150 + pembrolizumab (200 mg) combination therapy. Doses in the table are only shown for NDI-101150; [c]Includes both dose escalation and dose expansion cohorts; [d]Includes maculopapular rash.
TRAE, adverse event considered related to NDI-101150

Table 2. Any-grade IR-TRAEs occurring in ≥2 patients in the safety analysis set (N=88)[a,b]

TRAEs, n (% patients)	NDI-101150 monotherapy (n=76)					Combination therapy[c] (n=12)		
	50 mg (n=8)	100 mg[d] (n=36)	140 mg (n=5)	150 mg[d] (n=18)	200 mg (n=9)	50 mg (n=8)	100 mg (n=4)	Overall (n=88)
≥1 IR TRAE	3 (38)	5 (14)	1 (20)	2 (11)	5 (56)	2 (25)	1 (25)	19 (22)
Preferred term								
Diarrhea	0	1 (3)	0	1 (6)	3 (33)	0	1 (25)	6 (7)
Vomiting	0	0	0	2 (11)	2 (22)	0	0	4 (5)
Colitis	0	1 (3)	0	0	2 (22)	0	0	3 (3)
Nausea	0	0	0	2 (11)	1 (11)	0	0	3 (3)
Rash[e]	2 (25)	0	1 (20)	0	1 (11)	0	0	4 (5)
Abdominal pain	0	0	0	1 (6)	0	0	1 (25)	2 (2)
Constipation	0	1 (3)	0	0	0	0	1 (25)	2 (2)
Pneumonitis	1 (13)	1 (3)	0	0	0	0	0	2 (2)

[a]Patients reporting more than one event are counted only once for each preferred term; [b]IR-TRAEs were assessed by the investigator; [c]NDI-101150 + pembrolizumab (200 mg) combination therapy. Doses in the table are only shown for NDI-101150; [d]Includes both dose escalation and dose expansion cohorts; [e]Comprising rash (n=3) and maculopapular rash (n=1)
IR-TRAE, immune-related adverse event considered related to NDI-101150

Efficacy in patients with RCC receiving NDI-101150 monotherapy

- As of 12 August 2024, NDI-101150 monotherapy had resulted in objective responses (ORs) in three of 17 patients with RCC (18%), comprising a complete response (CR) and two partial responses (PRs) (**Fig. 3** and **4**, and **Table 3**)

Figure 3. Duration of treatment/BOR[a] in response-evaluable patients with RCC (N=17)[b]

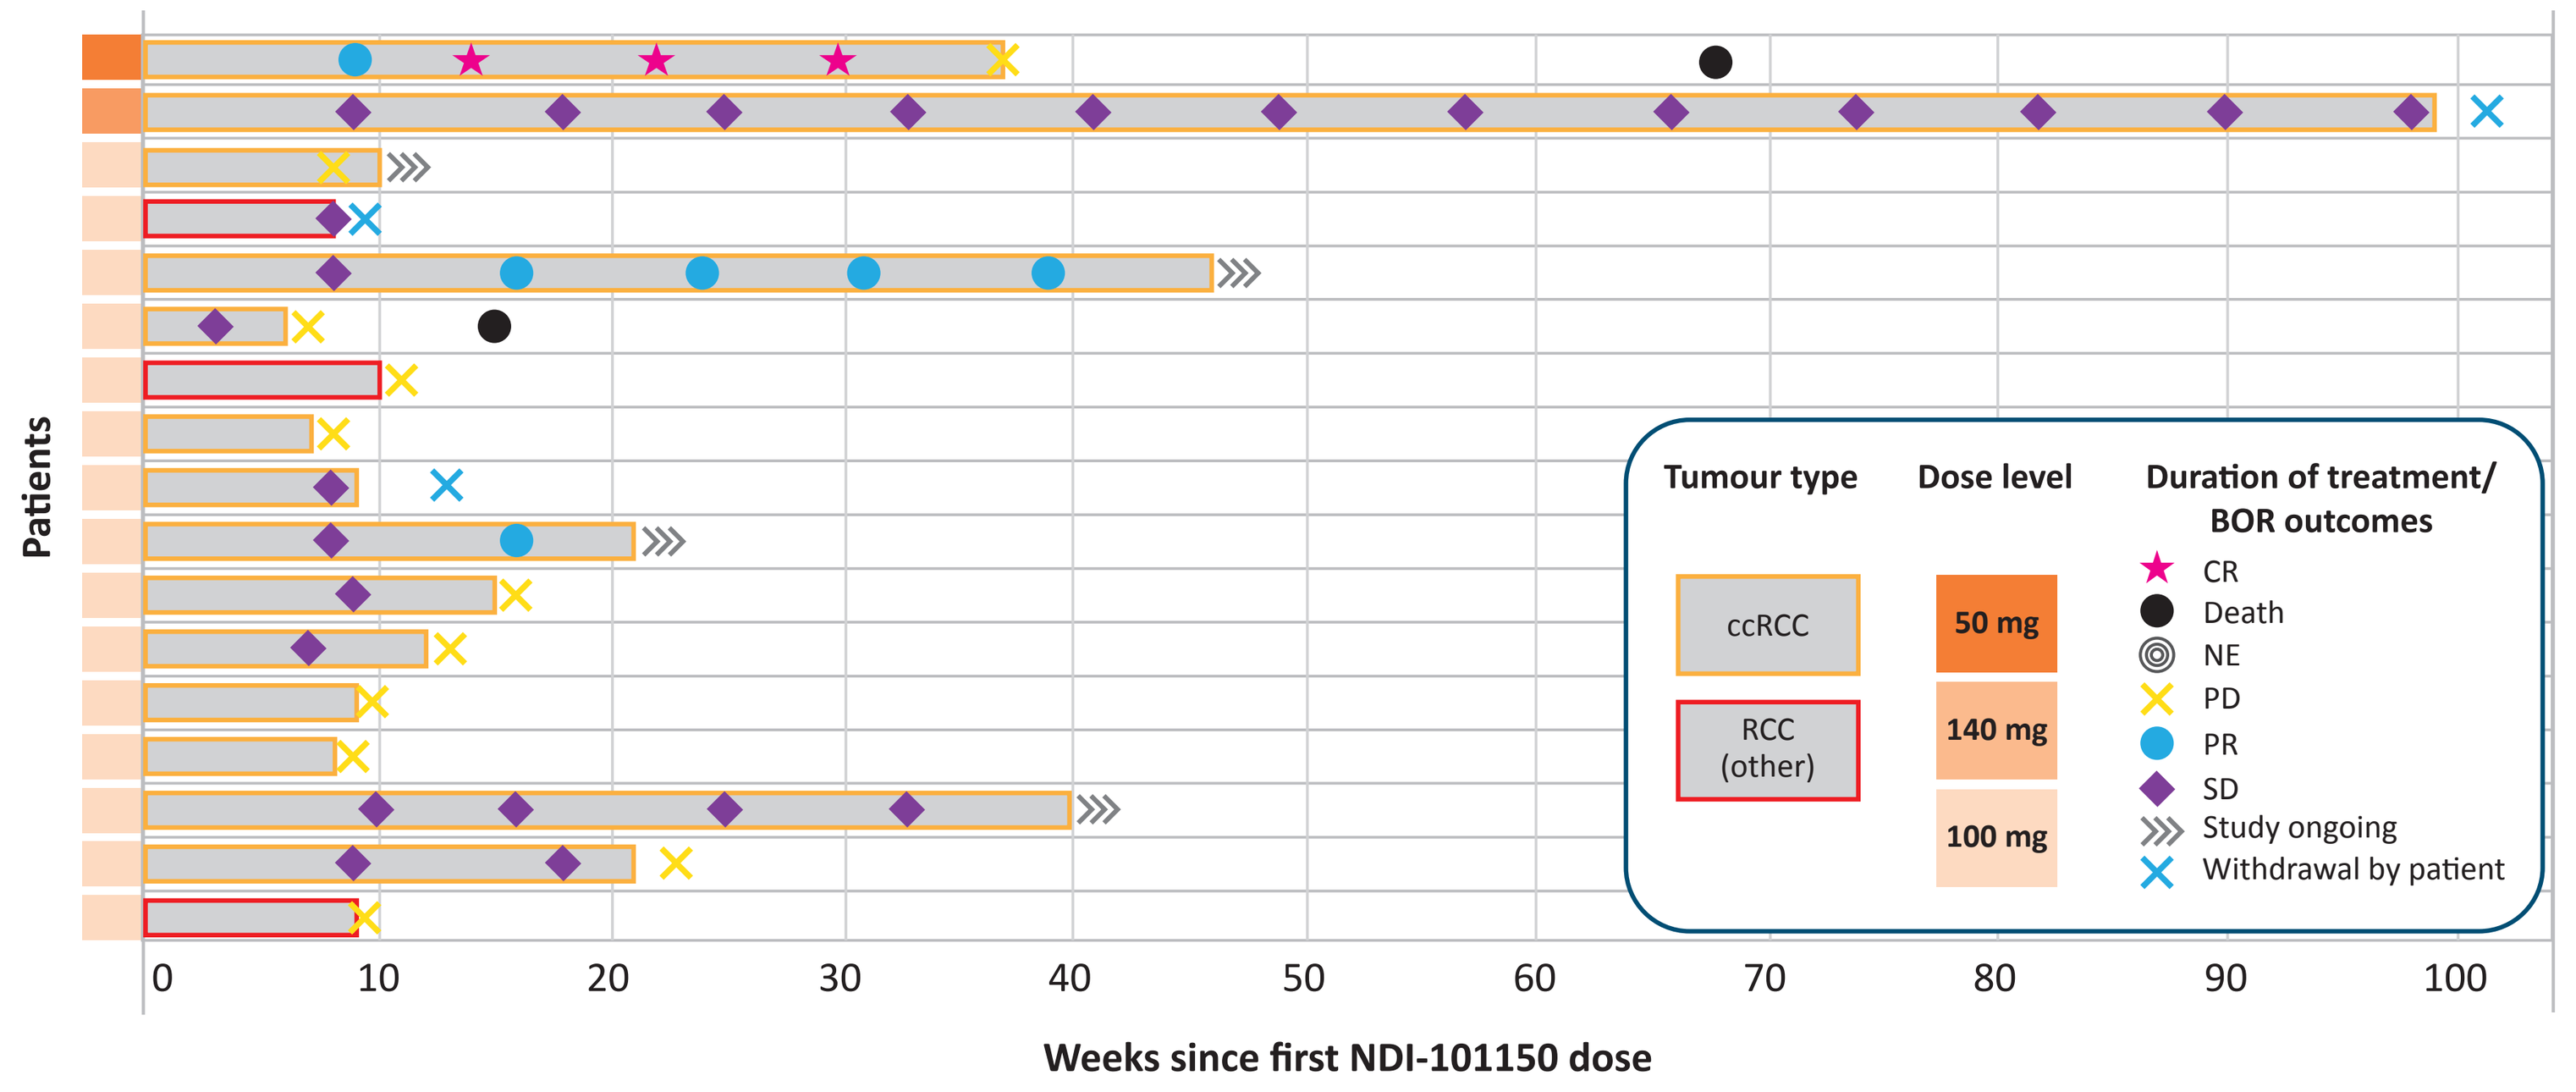

Patients
0
10
20
30
40
50
60
70
80
90
100
Weeks since first NDI-101150 dose
Tumour type
ccRCC
RCC (other)
Dose level
50 mg
140 mg
100 mg
Duration of treatment/ BOR outcomes
CR
Death
NE
PD
PR
SD
Study ongoing
Withdrawal by patient

[a]According to RECIST v1.1; [b]Analysis population included all patients with a post-baseline assessment
BOR, best overall response; ccRCC, clear cell renal cell carcinoma; CR, complete response; PD, progressive disease; PR, partial response; RCC, renal cell carcinoma; RECIST v1.1, Response Evaluation Criteria In Solid Tumors version 1.1; SD, stable disease

Table 3. BOR[a] in patients with RCC receiving NDI-101150 monotherapy (N=17)

BOR, n (%)	Patients with RCC (N=17)
ORR	3 (18)
CR	1 (6)
PR	2 (12)
SD	8 (47)
Durable SD[b]	2 (12)
PD	6 (35)

[a]According to RECIST v1.1; [b]≥9 months.
BOR, best overall response; CR, complete response; ORR, objective response rate; PD, progressive disease; PR, partial response; RCC, renal cell carcinoma; RECIST v1.1, Response Evaluation Criteria In Solid Tumors version 1.1; SD, stable disease.

- All patients with a confirmed CR or PR were heavily pretreated and had progressed on ≥1 prior checkpoint inhibitor (**Fig. 5A-C**)
- The clinical benefit rate (CR + PR + stable disease [SD] ≥6 months) was 29%; the disease control rate (CR + PR + any SD) was 65%

Figure 5. CT scans in a patient with ccRCC and a complete response (A), poor-risk ccRCC and a partial response (B) and ccRCC and a partial response (C)

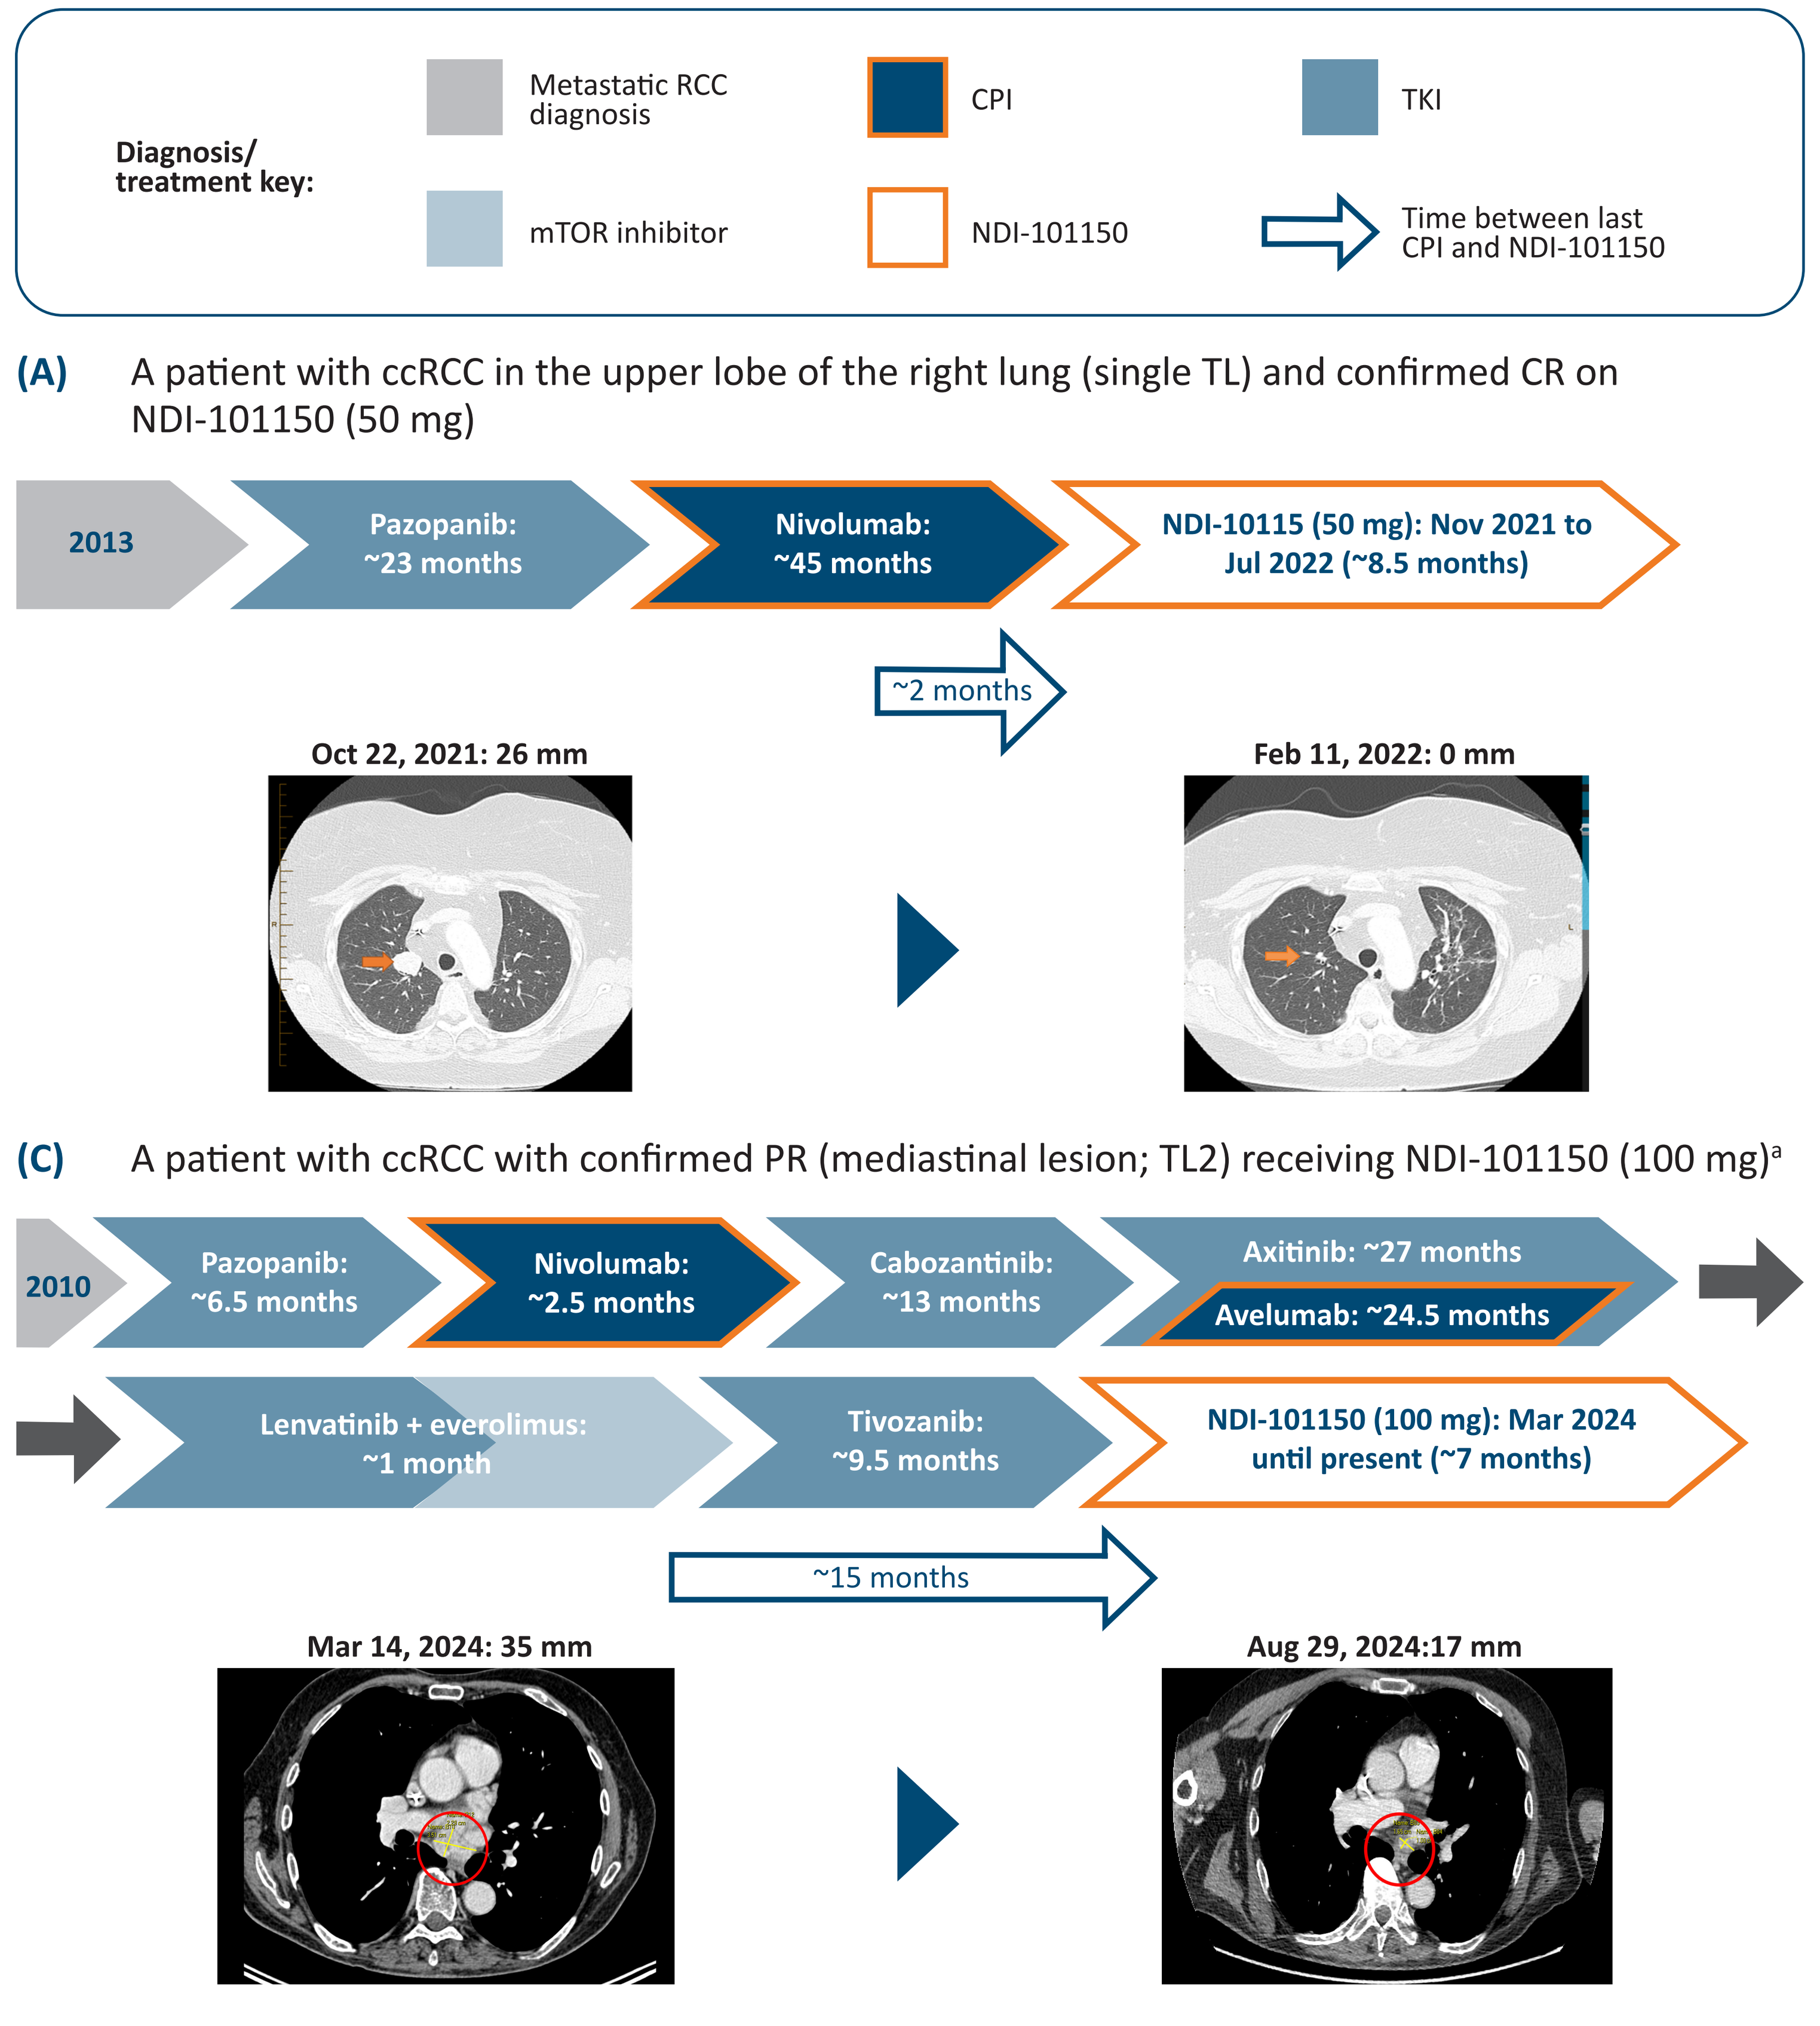

Diagnosis/ treatment key:
Metastatic RCC diagnosis
CPI
TKI
mTOR inhibitor
NDI-101150
Time between last CPI and NDI-101150
(A) A patient with ccRCC in the upper lobe of the right lung (single TL) and confirmed CR on NDI-101150 (50 mg)
2013
Pazopanib: ~23 months
Nivolumab: ~45 months
NDI-10115 (50 mg): Nov 2021 to Jul 2022 (~8.5 months)
~2 months
Oct 22, 2021: 26 mm
Feb 11, 2022: 0 mm
(C) A patient with ccRCC with confirmed PR (mediastinal lesion; TL2) receiving NDI-101150 (100 mg)[a]
2010
Pazopanib: ~6.5 months
Nivolumab: ~2.5 months
Cabozantinib: ~13 months
Axitinib: ~27 months
Avelumab: ~24.5 months
Lenvatinib + everolimus: ~1 month
Tivozanib: ~9.5 months
NDI-101150 (100 mg): Mar 2024 until present (~7 months)
~15 months
Mar 14, 2024: 35 mm
Aug 29, 2024:17 mm

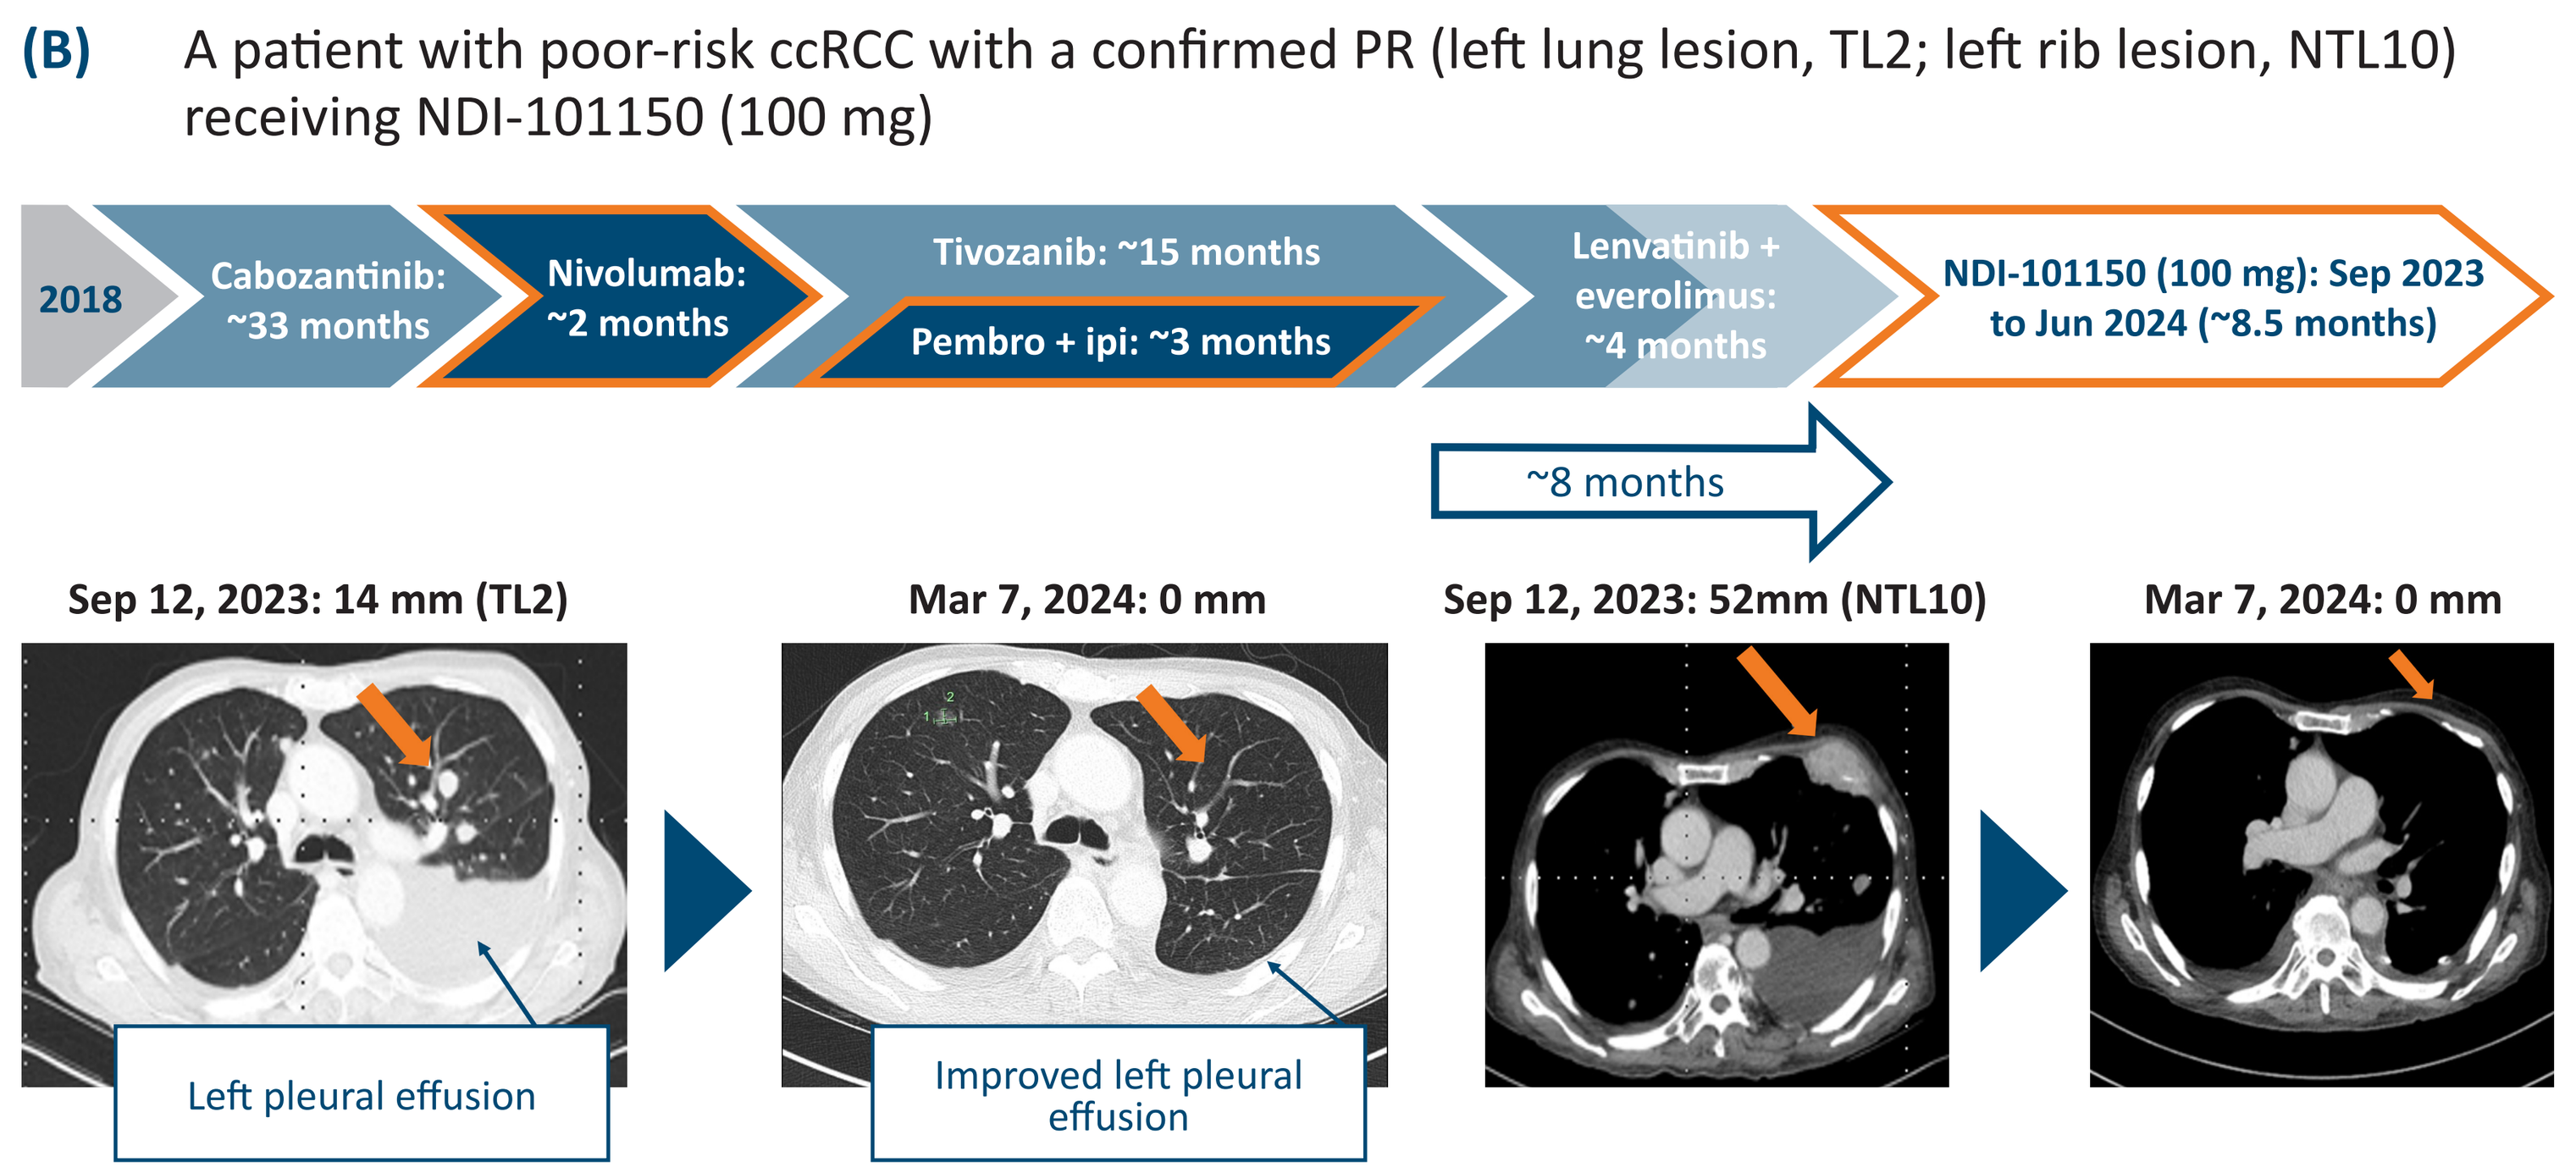

(B) A patient with poor-risk ccRCC with a confirmed PR (left lung lesion, TL2; left rib lesion, NTL10) receiving NDI-101150 (100 mg)
2018
Cabozantinib: ~33 months
Nivolumab: ~2 months
Tivozanib: ~15 months
Pembro + ipi: ~3 months
Lenvatinib + everolimus: ~4 months
NDI-101150 (100 mg): Sep 2023 to Jun 2024 (~8.5 months)
~8 months
Sep 12, 2023: 14 mm (TL2)
Mar 7, 2024: 0 mm
Sep 12, 2023: 52mm (NTL10)
Mar 7, 2024: 0 mm
Left pleural effusion
Improved left pleural effusion

[a]Patient also received a CD3 bispecific T cell engager (ENPP3xCD3) prior to NDI-101150 treatment
ccRCC, clear cell renal cell carcinoma; CPI, checkpoint inhibitor; CR, complete response; CT, computed tomography; ipi, ipilimumab; mTOR, mammalian target of rapamycin; NTL, non-target lesion; pembro, pembrolizumab; PR, partial response; TKI, tyrosine kinase inhibitor; TL, target lesion

Figure 4. Waterfall plot showing percent change in target lesion size by BOR in patients with RCC receiving NDI-101150 montherapy[a]

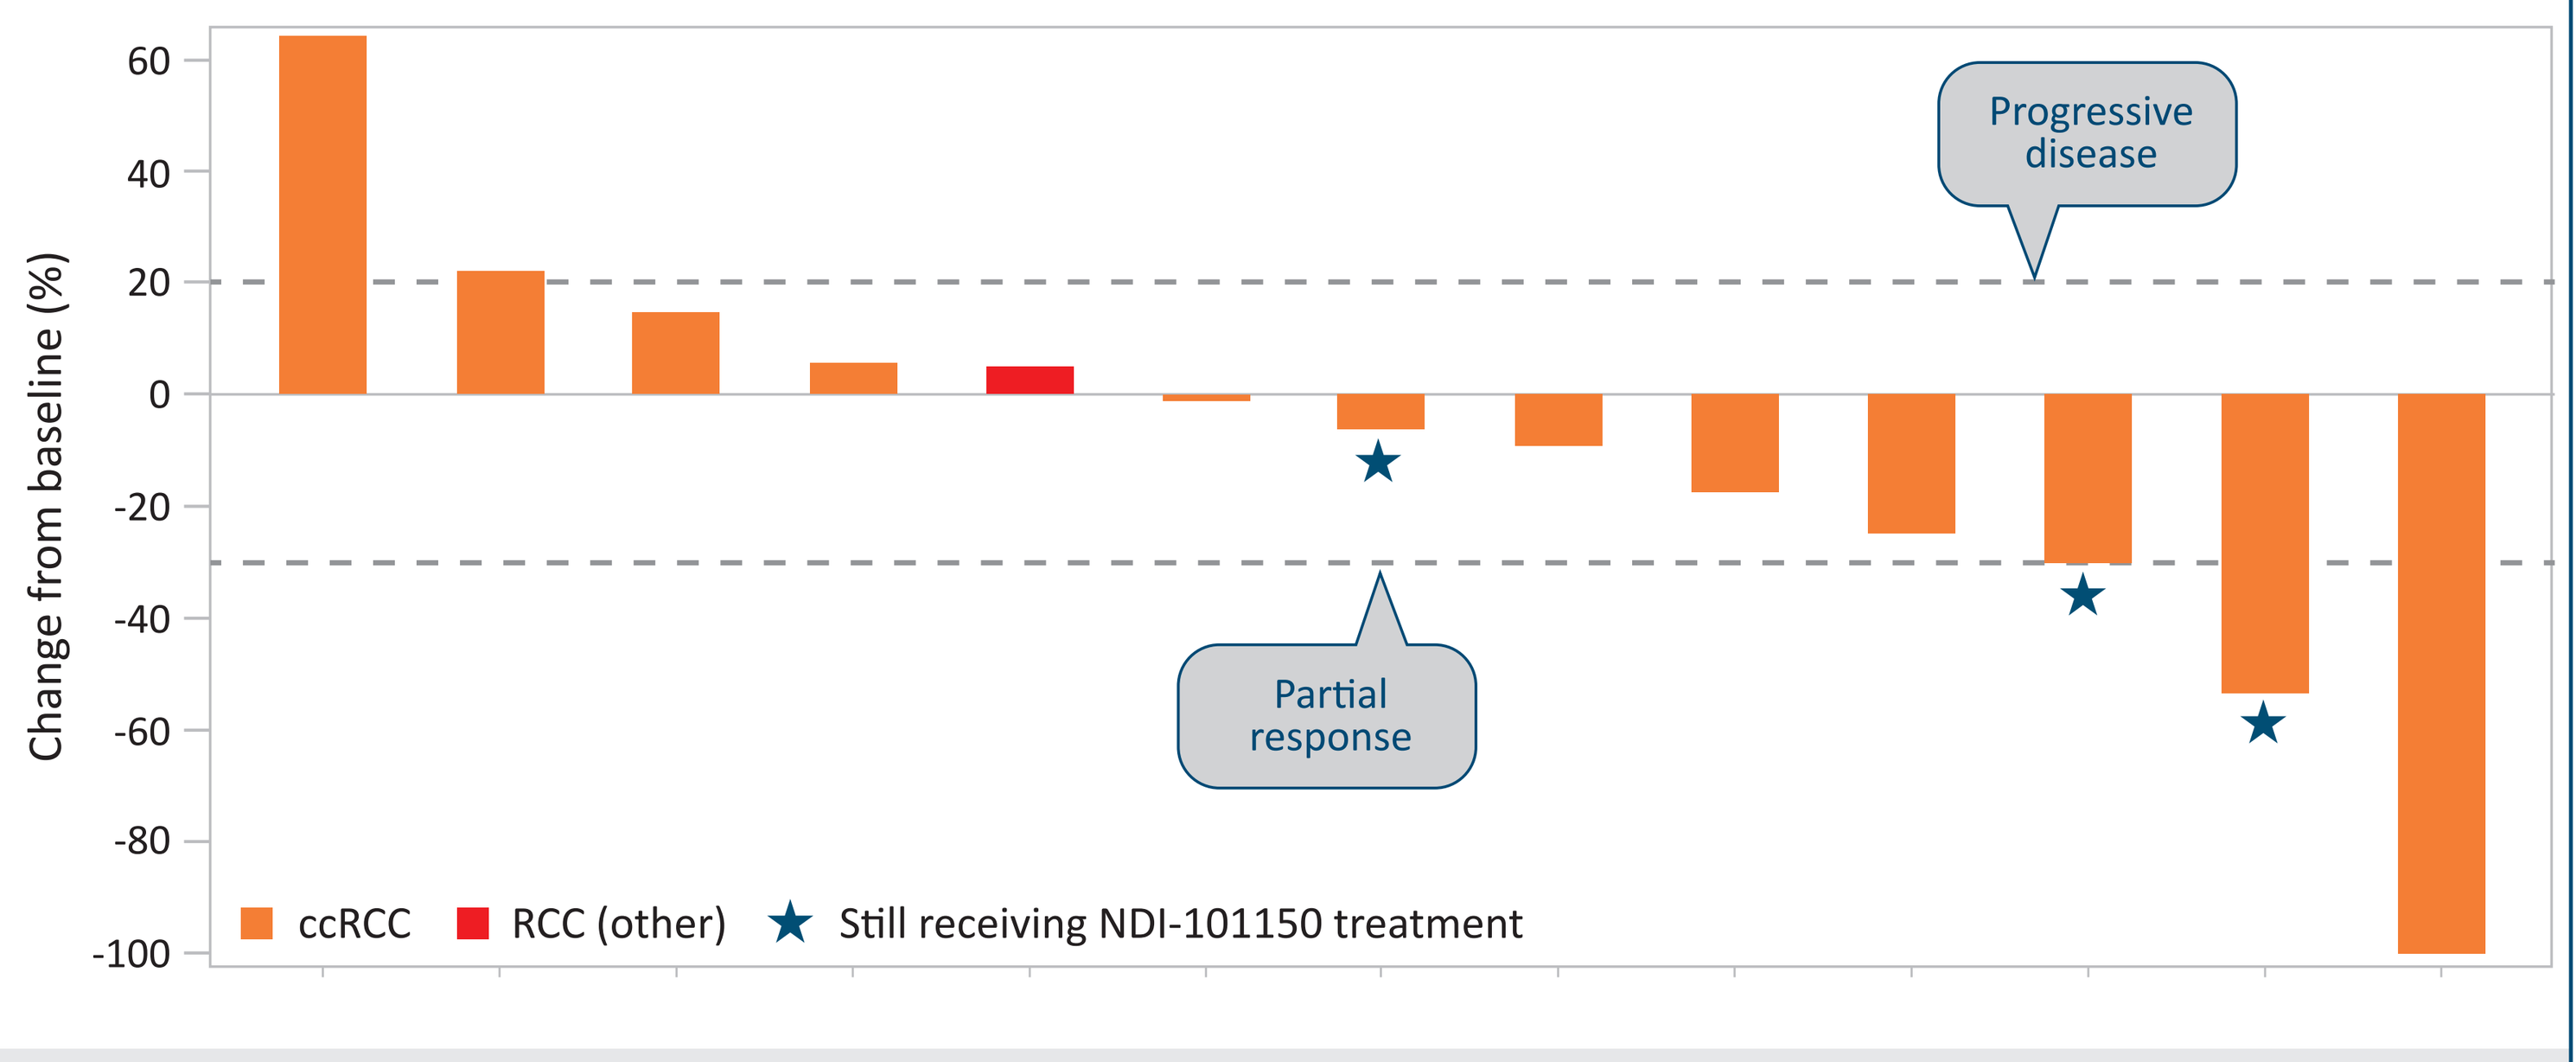

Change from baseline (%)
60
40
20
0
-20
-40
-60
-80
-100
Progressive disease
Partial response
ccRCC
RCC (other)
Still receiving NDI-101150 treatment

[a]Patients whose best response was PD due to new lesions, but whose target lesions had a BOR of SD or PR, were excluded
BOR, best overall response; ccRCC, clear cell renal cell carcinoma; RCC, renal cell carcinoma

CONCLUSIONS

- Objective responses were reported in three of 17 response-evaluable patients with RCC: one patient with a complete response and two patients with a partial response
- Clinical benefit (CR + PR + SD ≥6 months) was observed in five of 17 (29%) response-evaluable patients with RCC, including two patients with durable stable disease: ~22 months in one patient and 9 months in one patient still on treatment; the disease control rate was 65%
- NDI-101150 was generally well tolerated
- NDI-101150 continues to demonstrate encouraging antitumor activity in patients with RCC and an acceptable safety profile, supporting continued clinical evaluation of NDI-101150 (NCT05128487) as a promising next-generation immunotherapy small molecule
- Additional translational data, including results demonstrating robust target engagement and proof of biology, are being presented separately (SITC 2024 Poster 83)[4]

References: 1. Ciccone, D., et al. Poster C065, EORTC-NCI-AACR 2023; 2. Ciccone, D., et al. Poster 1340, SITC 2023; 3. Sommerhalder D, et al. Poster 751, SITC 2023; 4. Daigle S, et al. Poster 83; SITC 2024.

Acknowledgements: Editorial assistance was provided by Melody Watson, Bioscript Group, Macclesfield, UK, and supported by Nimbus Therapeutics (Nimbus Discovery Inc. on behalf of Nimbus Saturn Inc.). This study was funded by Nimbus Therapeutics (Nimbus Discovery Inc. on behalf of Nimbus Saturn Inc.).

Disclosures: **DS** has received honoraria from Syneos, consulting fees from Guidepoint and ongoing or past research funding from AbbVie, ADC Therapeutics, Ascentage Pharma Group, Astellas, Biomea Fusion, Boehringer Ingelheim, BJ Bioscience, BioNTech, Fate Therapeutics, Gilead Sciences, Haihe Pharmaceutical, IconOVir Bio, Immuneering, IMPACT Therapeutics, Kura Oncology, MediLink Therapeutics, Mirati Therapeutics, Monopteros Therapeutics, Navire Pharma Inc., Nimbus Saturn Inc., NGM Biopharmaceuticals, OncoResponse Inc., Parthenon, Pfizer, Revolution Medicines, Symphogen, Tachyon Therapeutics, Teon Therapeutics, Vincerx Pharma, ZielBio Inc.; **KD** has no relationships to disclose; **MSN** has held a consulting or advisory role for Celgene, Ipsen, and Taiho Pharmaceutical, has taken part in speakers' bureaus for Celgene, Daiichi Sankyo/Astra Zeneca, and Taiho Pharmaceutical, has received research funding from ERYTECH Pharma, and has received consulting fees from Pfizer; **HE** has held an advisory board role for BMS, Eisai, Aveo, Janssen and Cardinal Health; **JP** owns stock in zogen and his institution has received research funding from eFFECTOR Therapeutics, Kechow Pharma, Sermonix Pharmaceuticals, Novartis, ImmunityBio, Incyte, AbbVie, Macrogenics, Dizal Pharma, BerGenBio, Epizyme, Astellas Pharma, Janssen, AstraZeneca, Systimmune, QSAM, OncoC4 and Seagen; **RB** has received honoraria from Cardinal Health, has held a consulting or advisory role for Bristol Myers Squibb, and has received research funding from Bristol Myers Squibb; **RHM** has held a consulting/advisory board role for Puretech Health and IDEAYA Biosciences, and has received research funding from Nimbus Therapeutics and Repare Therapeutics; **AS**, **JH**, **DC**, **PK**, **SD**, **SD**, and **XY** are employees and equity holders of Nimbus Therapeutics (Nimbus Discovery Inc.) on behalf of Nimbus Saturn Inc; **MG** has received consulting fees from Guardant, Cellularity, and MSD, and has taken part in a data safety monitoring board/advisory board for Sanofi; **SS** has no relationships to disclose; **AH** has no relationships to disclose.

Presented at the Society for Immunotherapy of Cancer Annual Meeting | November 6–10, 2024 | Houston, TX, USA

Supported by Nimbus Therapeutics, Boston, MA